Poster # 846

19th Conference on Retroviruses and Opportunistic Infections. Seattle. March 5-8 2012

Correspondence: jibernardino.hulp@salud.madrid.org

Lopinavir-ritonavir plus Abacavir and Lamivudine versus Lopinavir-ritonavir Monotherapy for Recovery of Lipoatrophy in HIV-infected Patients with Sustained Virological Suppression while Receiving Zidovudine/Lamivudine/Abacavir

Bernardino JI[1], Pulido F[2], Martinez E[3], Arrizabalaga J[4], Domingo P[5], Portilla J[6], Ocampo A[7], Muñoz J[8], Torres, R[9], Arribas JR[1] for the GESIDA-6008-KRETA Study Group.

(1)Hospital Universitario La Paz, IdiPAZ, Madrid, Spain. (2)Hospital Universitario 12 de Octubre, Madrid, Spain. (3)Hospital Clínic, Institut d´Investigacions Biomediques August Pi i Sunyer, University of Barcelona, Spain. (4)Hospital Donostia, San Sebastián, Spain. (5)Hospital de la Santa Creu i San Pau. University of Barcelona, Spain. (6)Hospital General de Alicante, Alicante, Spain. (7)Complexo Hospitalario Universitario de Vigo, Pontevedra, Spain. (8)Hospital de Basurto, Bilbao, Spain. (9)Hospital Severo Ochoa, Leganes, Madrid, Spain.

BACKGROUND

- Prevalence of lipoatrophy varies from 50% in early cohort studies to 25% in recent studied cohorts[1,2].
- Thymidine analogues (both d4T and AZT) have been clearly associated with lipoatrophy[3].
- Discontinuation of thymidine nucleoside reverse transcriptase inhibitors (NRTIs) is the only proven strategy for lipoatrophy based on antiretroviral regimen change[4-6].
- Clinical trials of nuke-sparing regimens have shown similar increases in limb fat but with worsening dyslipidemia[7].
- Four studies in different scenarios suggest a role of protease inhibitors (PI) monotherapy for limb fat recovery[8-11].

HYPOTHESIS

Switching from ZDV/3TC/ABC to LPV/r monotherapy would result in more limb fat recovery than ABC/3TC + LPV/r in virologically suppressed HIV-infected patients with moderate to severe lipoatrophy

OBJECTIVES

Primary objective:

- Absolute change in limb fat mass measured by DEXA from baseline to W48

Secondary objectives:

- Absolute change in limb fat at 96 weeks from baseline
- Percentage change in limb fat mass at 48/96 weeks from baseline
- Changes in lipid profiles (Total, HDL and LDL cholesterol, triglycerides) at 24, 48 and 96 weeks
- Safety and tolerability
- Incidence of virologic failures (>400 HIV-RNA copies/mL)

KRETA STUDY DESIGN

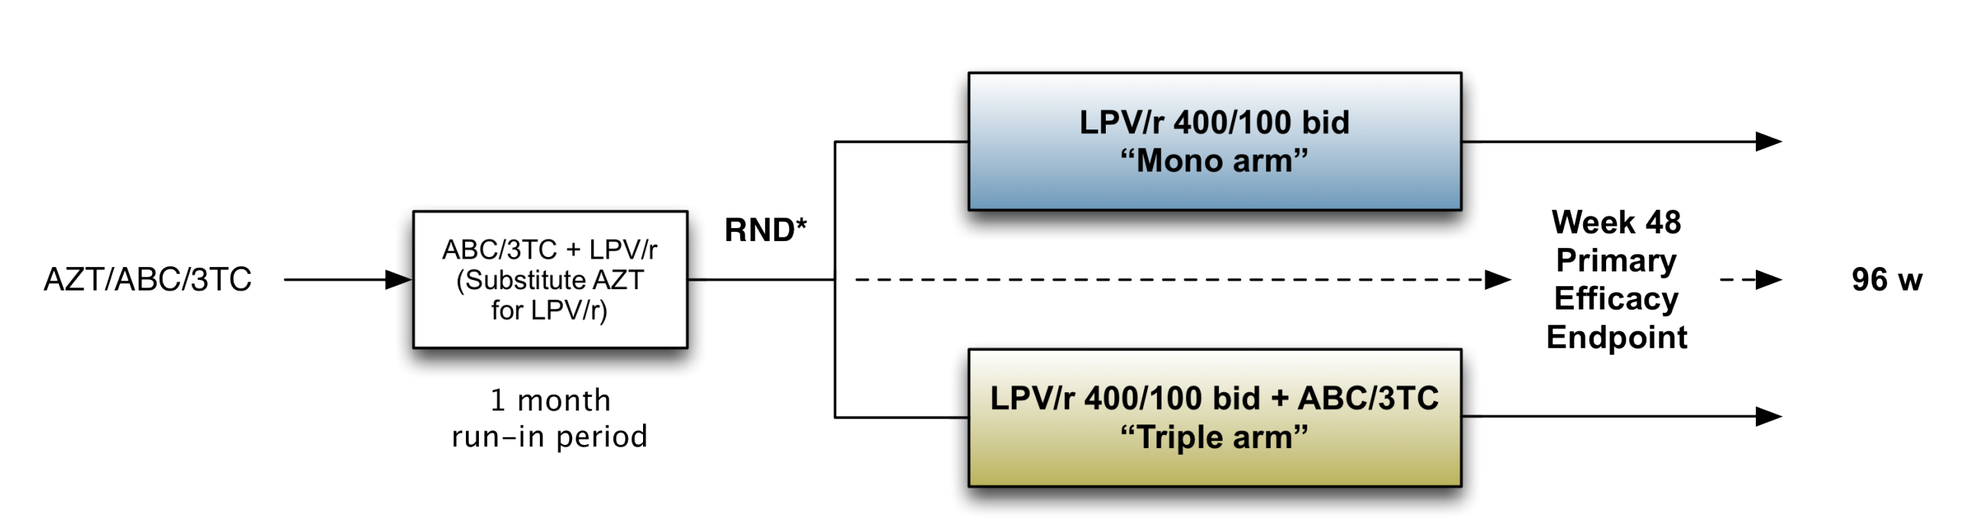
AZT/ABC/3TC
ABC/3TC + LPV/r (Substitute AZT for LPV/r)
1 month run-in period
RND*
LPV/r 400/100 bid "Mono arm"
LPV/r 400/100 bid + ABC/3TC "Triple arm"
Week 48 Primary Efficacy Endpoint
96 w

* RND: Randomization

Stratified by:	
Nadir CD4 cell count (< or ≥ 100 cells/mm3) Months with zidovudine (< or ≥ 3 years). DEXA center	
Inclusion criteria:	**Exclusion criteria:**
Stable Trizivir® (AZT/3TC/ABC) Moderate-severe lipoatrophy (LSGS) VL < 50 copies/ml for at least 6 month No prior virological failure of a PI containing regimen	Pregnancy Presence of serum hepatitis B surface antigen Chemotherapy for malignancy Insulin-sensitizing agent, anabolic steroids or GH in the last 16 weeks

METHODS

- 96 week multicentre, prospective, open label, randomized (1:1) study
- 10 sites in Spain
 - » Visits: Baseline, 4, 12, 24, 36, 48, 72, 96 weeks
 - » DEXAs: Baseline, 48, 96 week.
 - » Severe-Moderate Lipoatrophy (LSGS* grade 2-3 in at least 2 localizations)
- Primary Endpoint:
 - » Absolute change in limb fat mass measured by DEXA scan from baseline to 48 weeks

*** Lichtenstein KA, et al. Clinical assessment of HIV-associated lipodystrophy in an ambulatory population. Aids 2001;15(11):1389-1398**

SUBJECT DISPOSITION

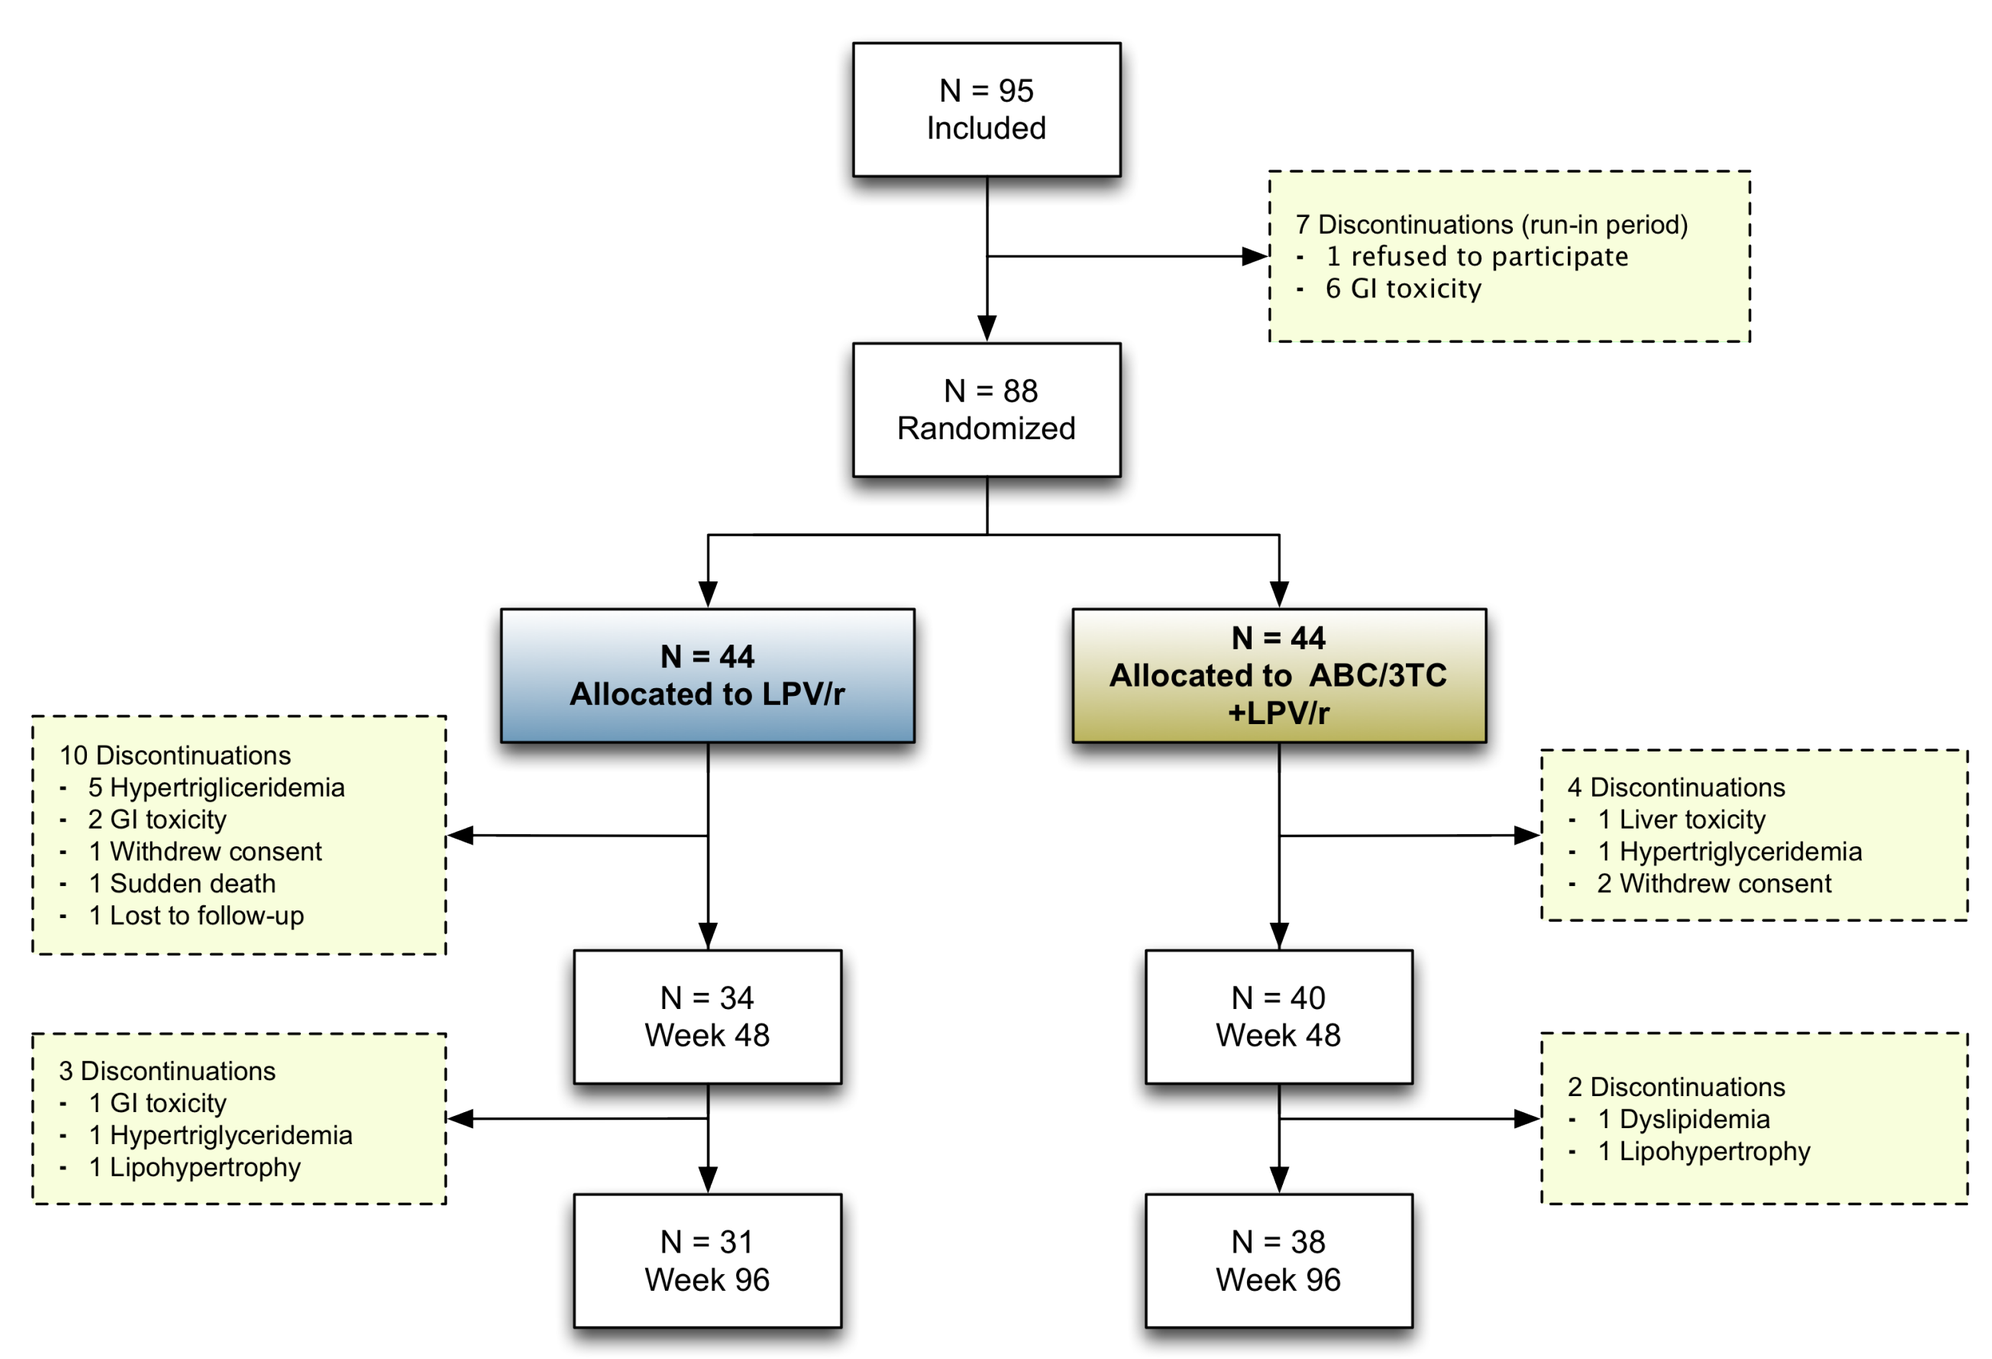
N = 95
Included
7 Discontinuations (run-in period)
- 1 refused to participate
- 6 GI toxicity
N = 88
Randomized
N = 44
Allocated to LPV/r
N = 44
Allocated to ABC/3TC +LPV/r
10 Discontinuations
- 5 Hypertriglyceridemia
- 2 GI toxicity
- 1 Withdrew consent
- 1 Sudden death
- 1 Lost to follow-up
4 Discontinuations
- 1 Liver toxicity
- 1 Hypertriglyceridemia
- 2 Withdrew consent
N = 34
Week 48
N = 40
Week 48
3 Discontinuations
- 1 GI toxicity
- 1 Hypertriglyceridemia
- 1 Lipohypertrophy
2 Discontinuations
- 1 Dyslipidemia
- 1 Lipohypertrophy
N = 31
Week 96
N = 38
Week 96

BASELINE: DEMOGRAPHICS AND CLINICAL CHARACTERISTICS

	Mono arm N = 44	Triple arm N = 44	Total N = 88	P value
Age [years (IQR)]	44.7 (41.5-52)	45 (42.1-50.8)	44.8 (41.8-51.1)	NS
Male sex [No. (%)]	26 (59.1)	33 (75)	59 (67)	NS
Mode of HIV transmission [No. (%)]				
Men sex with men	8 (18.2)	17 (38.6)	25 (28.4)	0.03
Heterosexual	22 (50)	14 (31.8)	36 (40.9)	0.08
IDU	11 (25)	15 (3 4)	26 (29.5)	NS
Other	3 (6.8)	2 (4.5)	5 (5.7)	NS
Duration HIV infection [years (IQR)]	11 (8.7-13.7)	11.8 (10.5-16.7)	11.4 (9.3-15)	0.09
CDC category AIDS [No. (%)]	26 (59.1)	20 (45.5)	46 (52.3)	NS
HCV + Antibody [No. (%)]	16 (36.4)	22 (50)	38 (43.2)	NS
Nadir CD4+ [cells/µL (IQR)]	189 (40-298)	233 (128-285)	222 (104-291)	NS
Total CD4+ [cells/µL (IQR)]	675 (512-885)	766 (543-1002)	697 (524-946)	NS
Median time on Zidovudine [years (IQR)]	6.7 (5.2-7.9)	8.5 (6.4-11.3)	7.4 (5.4-9.2)	0.003
Median time on thymidine analogues [years (IQR)]	7.9 (5.5-10.3)	10.5 (7.7-11.7)	9.4 (6.3-11)	0.006

BASELINE CHARACTERISTICS: ANTHROPOMETRICS, BODY COMPOSITION, LIPIDS

	Mono arm N = 44	Triple arm N = 44	Total N = 88	P value
Body mass index [Kg/m2 (IQR)]	24 (20.9-25.8)	23.3 (21-25)	23.5 (21-25)	NS
Waist circumference [cm (IQR)]	83.5 (75.9-91.3)	86 (80-91.5)	85 (78-91.5)	NS
Hip circumference [cm (IQR)]	91 (87-95.5)	89.5 (86-96)	90 (86-96)	NS
Total body fat [Kg (IQR)]	12.9 (8.9-16.5)	9.8 (7.4-15.4)	11.6 (8-15.7)	NS
Trunk fat [Kg (IQR)]	8.5 (5.6-11.1)	8.2 (5.1-10.8)	8.4 (5.4-10.8)	NS
Limb fat [Kg (IQR)]	2.5 (1.9-5.3)	2.5 (1.6-3.6)	2.5 (1.7-4.2)	NS
Fat mass ratio [median (IQR)]	1.9 (1.4-2.9)	2.1 (1.6-3.1)	2.0 (1.5-3.1)	NS
Total Cholesterol [mg/dL (IQR)]	247.5 (195-268)	214.2 (179.5-263)	224 (187-268)	0.075
HDL- Cholesterol [mg/dL (IQR)]	43.8 (38-58.5)	40 (36-49.5)	41.5 (37-53)	NS
LDL-Cholesterol [mg/dL (IQR)]	136 (98-167)	112 (86-142)	120 (94-160)	NS
Triglycerides [mg/dL (IQR)]	270 (156-398)	246 (134-342)	254 (151-378)	NS
Total /HDL-Cholesterol ratio [median (IQR)]	5.3 (4.1-6.7)	5.2 (3.8-6.3)	5.2 (3.9-6.5)	NS

MEAN CHANGE IN TOTAL, TRUNK, LIMB FAT AND FAT MASS RATIO

	Mean (SD) change over 48 weeks				Mean (SD) change over 96 weeks			
	Mono arm N =34	Triple arm N =40	Mean (95% CI) difference between arms*	P value	Mono arm N=32	Triple arm N=39	Mean (95% CI) difference between arms*	P value
Total fat (gr)	98.5 (3814)	394.6 (3628)	- 296 (-2032, 1440)	0.73	394 (4243)	419 (3830)	-25 (-1961, 1911)	0.98
Limb fat (gr)	215 (1161)	324.2 (1205)	- 109 (-659, 440)	0.69	416 (1670)	358.5 (1207)	57 (-650, 765)	0.87
Trunk fat (gr)	- 84 (2737)	110 (2533)	-194 (-1425, 1037)	0.75	-33 (2762)	64.5 (2601)	-98 (-1380, 1184)	0.88
Fat mass ratio †	-1 (0.5)	-0.5 (2.1)	0.42 (-0.28, 1.12)	0.23	-0.2 (0.5)	-0.6 (1.9)	0.4 (-0.23, 1.04)	0.20
Limb fat (%)	10 (44.6)	25 (64.6)	- 14.8 (-40.2, 10.7)	0.25	15.7 (62.7)	30.1 (72.9)	- 14.4 (-46.5, 17.7)	0.37

*** mean value in LPV/r minus ABC+3TC+LPV/r**
† Statistically significant difference intra-group for FMR in Mono arm at 96 w: -0.2 (95% CI -0.37, -0.02); p =0.026

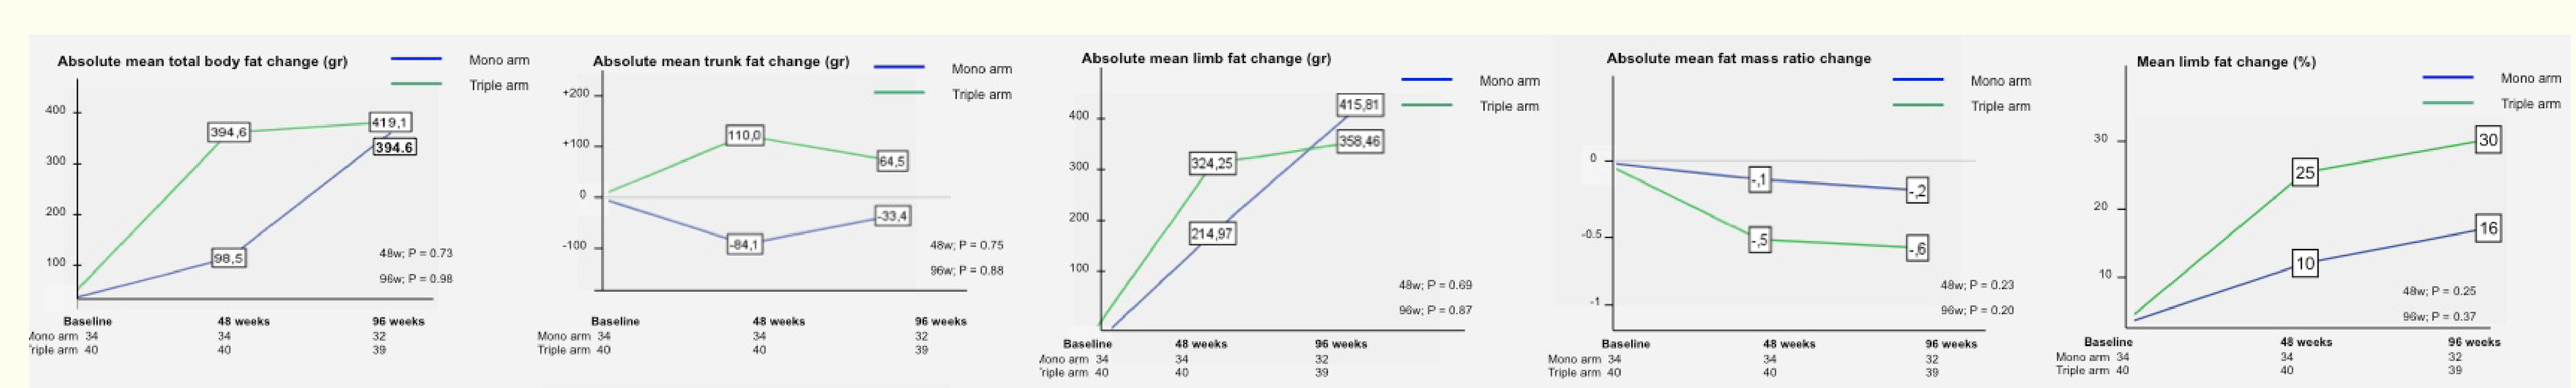
Absolute mean total body fat change (gr)
Absolute mean trunk fat change (gr)
Absolute mean limb fat change (gr)
Absolute mean fat mass ratio change
Mean limb fat change (%)
Mono arm
Triple arm
Baseline
48 weeks
96 weeks

LIMB FAT GAIN < 10%, 10-20%, 20-30%, >30%

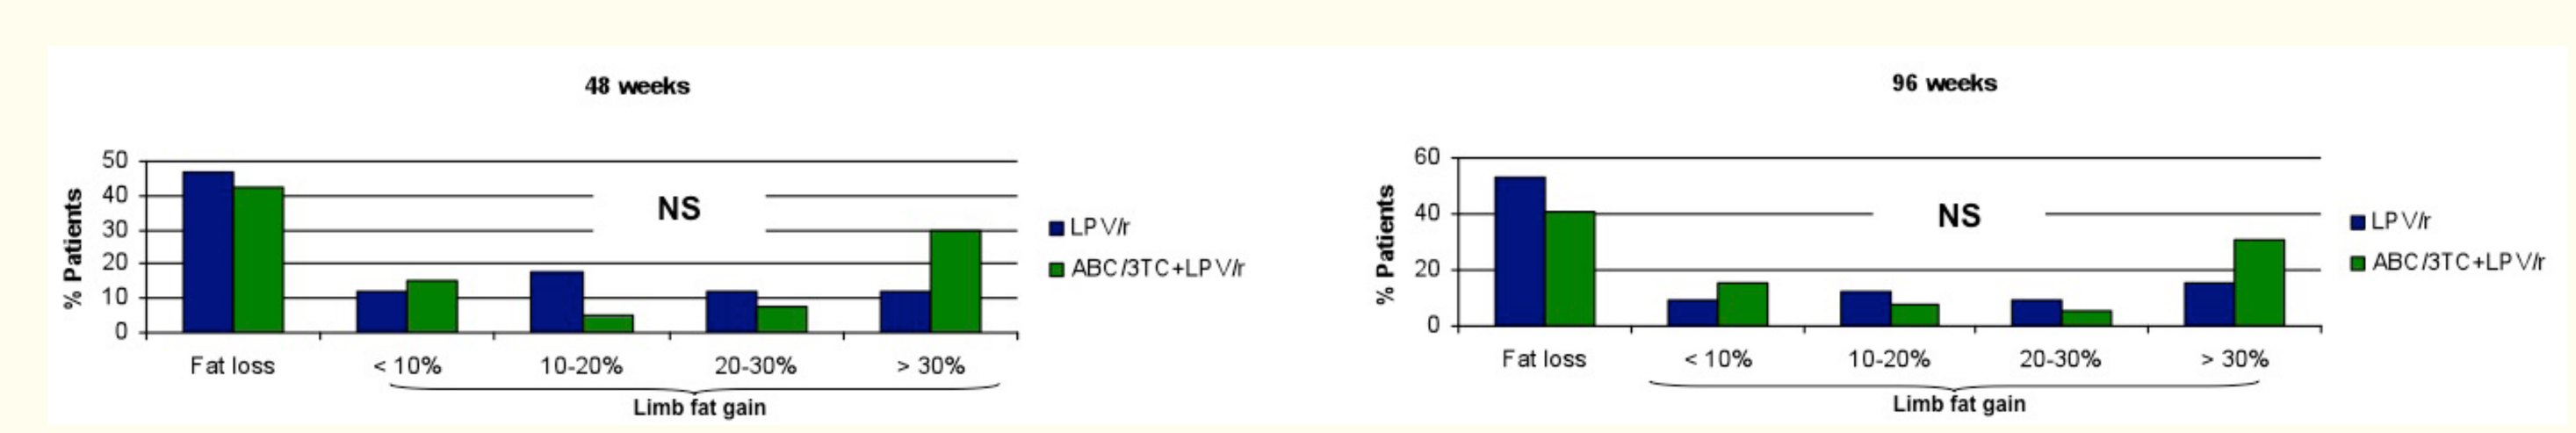
48 weeks
96 weeks
% Patients
NS
LPV/r
ABC/3TC+LPV/r
Fat loss
< 10%
10-20%
20-30%
> 30%
Limb fat gain

LIPID CHANGES FROM BASELINE TO WEEK 48 AND 96

	Mean (SD) change run-in period	Mean (SD) change from BL to W48				Mean (SD) change from BL to W96			
	N =87	Mono arm N =44	Triple arm N =43	Mean (95% CI) difference between arms*	P value	Mono arm N =44	Triple arm N =43	Mean (95% CI) difference between arms*	P value
Total Cholesterol (mg/dl)	44 (38)	- 7 (36)	10.2 (46)	-17.1 (-34.7, 0.49)	0.057	-8.7 (38.8)	4.7 (44)	-13.37 (-31, 4.3)	0.136
HDL cholesterol (mg/dl) †	-1.9 (9.4)	5 (10.5)	-0.6 (19.2)	5.6 (-1.02, 12.28)	0.096	6.6 (12.1)	2.7 (23.2)	3.8 (-4.1, 11.8)	0.342
LDL cholesterol (mg/dl)	19.4 (31.6)	0.3 (30.4)	16.6 (41)	-16.3 (-32.74, 0.11)	0.052	-3.4 (28.6)	8.2 (44)	-11.6 (-28.6, 5.4)	0.178
Triglycerides (mg/dl)**	151.6 (202)	- 74 (149)	-13 (129)	-61 (-121.5, 0.57)	0.048	-45.6 (261)	-27.6 (172.7)	-18 (-114.2, 78.1)	0.711
Total/HDL cholesterol †	1 (1.1)	- 0.5 (1.1)	0.3 (1.5)	-0.81 (-1.38, 0.23)	0.006	- 0.6 (1.4)	0.0 (2)	-0.62 (-1.37, 0.13)	0.104

*** mean value in LPV/r minus ABC+3TC+LPV/r**
† Statistically significant difference intra-group in HDL cholesterol and total/HDL cholesterol in mono arm at 48 w and 96 w:
HDL cholesterol: 48 week [5 (95% CI 1.7, 8.1); p =0.03], 96 week [6.5 (95% CI 2.8, 10.2); p =0.01]
Total/HDL cholesterol: 48 week [-0.53 (95% CI -0.85, -0.21); p =0.002], 96 week [-0.6 (95% CI -1.02, -0.17); p =0.007]
**** Statistically significant difference intra-group in triglycerides in mono arm at 48w: -74 (95% CI -119.5, -28.6); p =0.002**

VIROLOGIC FAILURES

- 3 virologic failures (2 mono arm/ 1 triple arm).
- The 82A mutation was detected in one patient in the monotherapy arm

PERCENTAGES OF PATIENTS WITH ADVERSE EVENTS 96 WEEKS

	Mono arm (N=44) n (%)	Triple arm (N=44) n (%)	P value
Any Adverse Event	42 (95.4)	35 (79.5)	0.024
Drug related Adverse Event	30 (68.2)	30 (68.2)	1
Serious Adverse Event*	8 (18.2)	5 (11.4)	0.367
Discontinuation due to any Adverse Event †	11 (25)	4 (9)	0.047

*** Serious Adverse Events:**
Mono arm: 1 cardiac arrest, 4 hospitalizations due to infectious diseases, 2 traumatic fractures, 1 paranoid disorder
Triple arm: 2 hospitalizations due to infectious diseases, 1 pneumothorax, 1 uterine mioma and 1 abdominal obesity surgery
† Most AE related discontinuations occurred before W24. The difference between arms is statistically significant at W48

DISCUSSION

- In previous studies, switching thymidine analogues for tenofovir or abacavir led to a median increase of limb fat mass between 300-480 gr at 48 weeks [4,5].
- Switching studies with PI monotherapy showed a limb fat increase between 160-340 gr at 48 weeks [10,12].
- Although limb fat increases in this trial are concordant with previous studies we didn't find differences between switching to LPV/r monotherapy or ABC/3TC + LPV/r. This could be explained by the following:
 - » Median baseline limb fat extremely low compared to other similar studies.
 - » Compared to previous studies longer prior exposure to thymidine analogues.
- Limitations:
 - » DEXA scans were not centrally read and different equipments (Lunar, Hologic, Norland) were used.
 - » Small number of patients that lead to misbalanced baseline characteristics although after adjustment main results didn't change (data not shown).
 - » Excessive numbers of discontinuations specially in the monotherapy arm which had an impact on the virological efficacy analysis.

CONCLUSIONS

- In moderate to severe lipoatrophic patients treated with AZT/ABC/3TC, with a long history of of thymidine analogues treatment, switching to LPV/r monotherapy had no benefit in limb fat recovery relative to switching to ABC/3TC + LPV/r.
- There were no differences in limb fat gain and no clinically relevant differences in lipid profile between both strategies.
- There were more discontinuations in the LPV/r monotherapy arm during the first 48 weeks
- These data suggest that non-thymidine nucleosides such as ABC/3TC do not represent themselves an obstacle for limb fat recovery.

ACKNOWLEDGEMENTS

- We would like to thank the GESIDA-6008-KRETA Study participants.
- GESIDA-6008-KRETA study was supported by an unrestricted grant of Abbott laboratories.
- Herminia Esteban from SEIMC-GESIDA foundation on behalf of the coordinating centre staff.

REFERENCES

1. Carr A, et al. A syndrome of peripheral lipodystrophy, hyperlipidaemia and insulin resistance in patients receiving HIV protease inhibitors. Aids 1998;12:F51-58. 2. Nguyen A, et al. Lipodystrophy and weight changes: data from the Swiss HIV Cohort Study, 2000-2006. HIV Med 2008;9:142-150. 3. Mallal SA, Contribution of nucleoside analogue reverse transcriptase inhibitors to subcutaneous fat wasting in patients with HIV infection. Aids;14:1309-1316. 4. Moyle GJ, et al. A randomized comparative trial of tenofovir DF or abacavir as replacement for a thymidine analogue in persons with lipoatrophy. Aids 2006;20:2043-2050. 5. Fisher M, et al. A randomized comparative trial of continued zidovudine/lamivudine or replacement with tenofovir disoproxil fumarate/emtricitabine in efavirenz-treated HIV-1-infected individuals. J Acquir Immune Defic Syndr. 2009;51:562-568. 6. European (EACS) guidelines http://www.europeanaidsclinicalsociety.org/images/stories/EACS-Pdf/eacsguidelines-v6_english.pdf. 7. Tebas P, et al. Peripheral and visceral fat changes following a treatment switch to a non-thymidine analogue or a nucleoside-sparing regimen in HIV-infected subjects with peripheral lipoatrophy: results of ACTG A5110. J Antimicrob Chemother. 2009;63:998-1005. 8. Kolta S, et al. Fat tissue distribution changes in HIV-infected patients treated with lopinavir/ritonavir. Results of the MONARK trial. Curr HIV Res. 2011;9:31-39. 9. Cameron DW, et al. A 96-week comparison of lopinavir-ritonavir combination therapy followed by lopinavir-ritonavir monotherapy versus efavirenz combination therapy. J Infect Dis. 2008;198:234-240. 10. Valantin MA, et al. Fat tissue distribution changes in HIV-infected patients with viral suppression treated with darunavir/ritonavir (DRV/r) monotherapy versus 2 NRTI + DRV/r in the MONOI--ANRS 136 Randomized Trial : Results at 48 weeks. 17th Conference on Retroviruses and Opportunistic Infections. February 16-19, 2010. San Francisco. # 721. 11. Guaraldi et al. Impact of Switching to Darunavir/ritonavir monotherapy vs. triple-therapy on Body Fat Redistribution and Bone Mass in Virologically Suppressed HIV-Infected Adults. The MONARCH randomized controlled trial. 13th European AIDS Conference EACS, Belgrade, Serbia, October 2011 [PE7.5/4.]. 12 Maynard JL, et al. Lopinavir/ritonavir monotherapy versus current treatment continuation for maintenance therapy of HIV-1 infection: the KALESOLO trial. J Antimicrob Chemother 2010;65:2436-2444

FSG FUNDACIÓN SEIMC-GESIDA