P-174

mluisa.montes@salud.madrid.org

Prognostic Utility of Transient Elastography In HIV-Infected Patients With Liver Cirrhosis

ML Montes[1], M Von Wichmann[2], JM Miró[3], C Quereda[4], C Tural[5], E Ortega[6], J Berenguer[7], J Sanz[8], A Hernando[9], Jose Arribas[1], and GESIDA 37/03-FIPSE 36680/07 Study Group.

(1) Hosp La Paz, IdiPAZ, Madrid, Spain; (2) Hospital de Donostia, San Sebastián, (3) Hosp. Clinic-IDIBAPS, Univ. of Barcelona, Barcelona, Spain, (4) Hosp. Ramon y Cajal, Madrid, Spain, (5) Hospital Germans Trials y Pujol, Badalona, Spain; (6) Hospital Universitario General de Valencia, Spain; (7) Hospital Universitario Gregorio Marañón, Madrid, Spain, (8) H. Universitario Príncipe de Asturias, Madrid, Spain; (9) Hospital 12 de Octubre, Universidad Europea de Madrid, Madrid, Spain; and (88) Fundación SEIMC-GESIDA, Madrid, Spain.

INTRODUCTION

- Liver cirrhosis is an important cause of mortality in HIV-infected subjects.
- After the first hepatic decompensation survival decreases sharply.
- Early diagnosis of liver cirrhosis and optimal management are essential to decrease the risk of hepatic decompensations and to improve prognosis in HIV-infected subjects.
- Transient elastograhy (TE) has been incorporated to the routine management of HIV-infected subject with liver cirrhosis to measure liver stiffness (LS).
- TE beside the good performance for the diagnosis of liver cirrhosis is a sensitive test to detect changes in the liver damage and portal hypertension.
- It is not known if liver stiffness measurements could predict hepatic mortality in HIV-infected subjects with chronic hepatitis.

OBJECTIVE

- To assess the performance of TE to predict hepatic decompensation and mortality in HIV cirrhotic subjects.

PATIENTS AND METHODS

- From the GESIDA prospective cohort of HIV-infected patients with confirmed liver cirrhosis in Spain with follow-up between 2004-2012 we selected all subjects who had a TE measurement at least 6 month before the last visit.
- Combined primary endpoint: death or hepatocarcinoma or liver transplant.
- Episodes of hepatic decompensation at baseline and during follow-up were collected.
- We evaluated by univariate/multivariate Cox proportional hazard models the prognostic value to predict hepatic decompensation or a combined endpoint (death, hepatocarcinoma or liver transplant) of LS measurement, CD4+ (nadir, baseline and <200 at baseline), ART, history of anti-HCV treatment, sustained viral response (SVR) to anti-HCV treatment, concomitant chronic hepatitis B, current Child-Pugh score (CPS), MELD, FIB4 score, APRI score.
- The sensitivity, specificity, PPV, NPV, positive likelihood ratio (LH+) and negative likelihood ratio (LH-) were calculated. LS measurement cutoff point was selected using ROC curve.

RESULTS

- 160 subjects had one TE measurement with at least 6 months of follow-up later in the cohort. Baseline characterisitcs are referred to the characteristic collected in the visit when TE measurement was performed (table 1).
- The median of follow-up was 36 months. 19 primary outcomes and 21 hepatic decompensation occurred during follow-up (table 2).
- Variables associated to combined endpoint were analyzed through univariate and multivariate methods. Two different models of multivariate analysis were conducted including or excluding hepatic decompensation variable (table 3 and 4).
- TE measurement of 44 Kpa had the best diagnostic performance and was selected.
- Survival analysis showed significant differences in subjects with TE measurement higher than 44 kpa and hepatic decompensation at baseline (figures 1 and 2).
- TE measurement performance for predicting clinical outcomes are shown in table 5.
- TE measurement for combined endpoint and death AUROC had a high performance (figure 3 and 4).

BASELINE CHARACTERISTICS

Table 1

Mean age, years (SD)	45 (6.7)
Female (%)	39 (24.4)
Cirrhosis diagnosis	
- Biopsy (%)	127 (79.4)
- Bonacini Score >8 (%)	21 (13.1)
Cirrhosis etiology	
- Hepatitis C (%)	152 (95.6)
- Genotypes 2,3 (%)	41 (28.7)
- Hepatitis B (%)	12 (8.4)
- Prior alcohol abuse (%)	40 (25.0)
Child Pugh score	
- CP A (%)	83 (55)
- CP B, C (%)	67 (45)
Compensated liver cirrhosis (%)	138 (86.3)
Median duration HIV infection (years), (IQR)	17.8 (12.6- 20.8)
Median duration HCV infection (years) (IQR)	11.7 (8.6- 14.2)
Transmission route IVDU (%)	139 (86.9)
CDC stage C (%)	52 (32.7)
Receiving ART at baseline (%)	151 (94.4)
HIV RNA load < 50 copies/mL (%)	138 (86.8)
Median CD4 cell count (IQR)	
- Baseline	448 (303-642)
- Nadir	184 (69.5-268)
Previous therapy against HCV (%)	125 (82.2)
- Sustained virological response (%)	34 (27.2)
- Still receiving HCV treatment (%)	5 (4.0)
- Non responders or relapsers (%)	86 (68.8)
Median Alanine aminotransferase , IU/L (IQR)	51 (30.0-79.5)
Median Aspartate aminotransferase, IU/L (IQR)	49 (32.0-83)
Median Total bilirubin, mg/dL (IQR)	0.9 (0.5-1.6)
Median Platelet count, mm3 (IQR)	118 (81.0-182.0)
MELD score	
- < 14	82 (96.5)
- > 14	3 (3.5)
Median Liver stiffness, kPa (IQR)	16.9 (9.5-26.3)
Liver stiffness, kPa (%)	
- < 21	100 (62.5)
- 21-39.9	38 (23.8)
- > 40	22 (13.8)
APRI score	
- < 0.5	42 (26.6)
- 0.5-1.5	53 (33.5)
- > 1.5	63 (39.9)
FIB4 score	
- < 1.45	43 (27.4)
- 1.45-3.25	53 (33.8)
- > 3.25	61 (38.9)

RESULTS

Table 2: clinical events during follow-up

	N	%
Follow-up, median (IQR) months	36 (20-47)	-
Lost to follow-up	5	3
Endpoints,		
Any	19	12
Death	14	8.8
Hepatocarcinoma	6	3.8
Transplant	3	1.9
Deaths,		
Hepatic causes	8	5
Other	6	3.7
Unknown	0	-
Decompensation during follow-up,		13
First decompensation	13	8
New decompensation	8	5

VARIABLES ASSOCIATED TO COMBINED ENDPOINT

Table 3: model 1 excluding hepatic decompensation

	Univariate analysis HR (CI 95%)	P	Multivariate analysis	P
Age (cuantitative)	1.02 (0.94-1.11)	0.58	-	-
Female	1.06 (0.38-2.94)	0.91	-	-
HCV sustained virological response	0.17 (0.02-1.4)	0.10	-	-
Receiving ART at baseline	1,19 (0.16-8.9)	0.86	-	.
HIV RNA BLQ at baseline	0.75 (0.2-2.6)	0.65	-	-
CD4<200 cel/µL at baseline	5.8 (2.6-14.8)	<0.000	2.7 (1.01-7.5)	0.047
- Child Pugh score B,C at baseline	1.3 (0.5-3.3)	0.6	-	-
- Platelets (cuantitative)	0.98 (0.96-0.99)	0.001	ns	-
- Platelets <120.000 cel/mL	20.2(2.66-154)	0.004		
MELD > 14	0.05 (0-400)	0.66	-	-
APRI (cuantitative)	1.5 (1.25-1.8)	<0.000	ns	-
APRI > 1.5	5.5 (1.8-16)	0.003		
FIB4 (cuantitative)	1.35 (1.2-1.5)	0.000	-	-
FIB4 > 3.25	14.3 (3.2-62)	0.000	6.3 (1.2-32)	0.03
TE measure (cuantitative)	1.05 (1.03-1.07)	0.000		
TE measure > 44 Kpa	12 (4.6-32)	0.000	4.2 (1.5-12)	0.008

Table 4: model 2 including hepatic decompensation

	Univariate analysis HR (CI 95%)	P	Multivariate analysis	P
Age (cuantitative)	1.02 (0.94-1.11)	0.58	-	-
Female	1.06 (0.38-2.94)	0.91	-	-
HCV sustained virological response	0.17 (0.02-1.4)	0.10	-	-
Receiving ART at baseline	1,19 (0.16-8.9)	0.86	-	.
HIV RNA BLQ at baseline	0.75 (0.2-2.6)	0.65	-	-
CD4<200 cel/µL at baseline	5.8 (2.6-14.8)	<0.000	ns	-
- Child Pugh score B,C at baseline	1.3 (0.5-3.3)	0.6	-	-
- Platelets (cuantitative)	0.98 (0.96-0.99)	0.001	ns	-
- Platelets <120.000 cel/mL	20.2(2.66-154)	0.004		
Decompensation at baseline	9.4 (3.6 – 25)	<0.001	4.5 (1.6 -13)	0.005
MELD > 14	0.05 (0-400)	0.66	-	-
APRI (cuantitative)	1.5 (1.25-1.8)	<0.000	ns	-
APRI > 1.5	5.5 (1.8-16)	0.003		
FIB4 (cuantitative)	1.35 (1.2-1.5)	0.000	-	-
FIB4 > 3.25	14.3 (3.2-62)	0.000	8.2 (1.7-40)	0.009
TE measure (cuantitative)	1.05 (1.03-1.07)	0.000		
TE measure > 44 Kpa	12 (4.6-32)	0.000	3 (1.03-9)	0.04

VARIABLES ASSOCIATED TO HEPATIC DECOMPENSATION

Table 5

	Univariate analysis HR (CI 95%)	P	Multivariate analysis	P
Age (cuantitative)	1.02 (0.94-1.11)	0.58	-	-
Female	0.65 (0.22-1.93)	0.4	-	-
HCV sustained virological response	0.03 (0.00-6.2)	0.2	-	-
Receiving ART at baseline	0.25 (0.07-0.9)	0.03	-	.
HIV RNA BLQ at baseline	0.33 (0.1-0.9)	0.04	ns	-
Chronic hepatitis B	6.4 (1.2-35)	0.03	ns	
CD4<200 cel/µL at baseline	4.24 (1.6-11)	<0.003	2.8 (1.06-7.8)	0.04
- Child Pugh score B,C at baseline	0.67 (0.25-1.8)	0.4	-	-
- Platelets (cuantitative)	0.98 (0.97-1)	0.005	ns	-
- Platelets <120.000 cel/mL	5.46(1.6-19)	0.008	ns	-
Esophageal varices/PH*	3.6 (1.1-12)	0.03	-	ns
MELD > 14	0.05 (0-400)	0.66	-	-
APRI (cuantitative)	1.4 (1.2-1.7)	<0.001	ns	-
APRI > 1.5	4.9 (1.7-13)	0.002	ns	-
FIB4 (cuantitative)	1.3 (1.1-1.4)	<0.001	-	-
FIB4 > 3.25	9 (2.5-31)	0.001	4.7 (1.1-19)	0.032
TE measure (cuantitative)	1.04 (1.02-1.06)	<0.001		
TE measure > 44 Kpa	6.4 (2.4-17)	<0.001	3.6 (1.32-10)	0.01

* PH: portal hypertension

COMBINED ENDPOINT AND LS MESAUREMENT

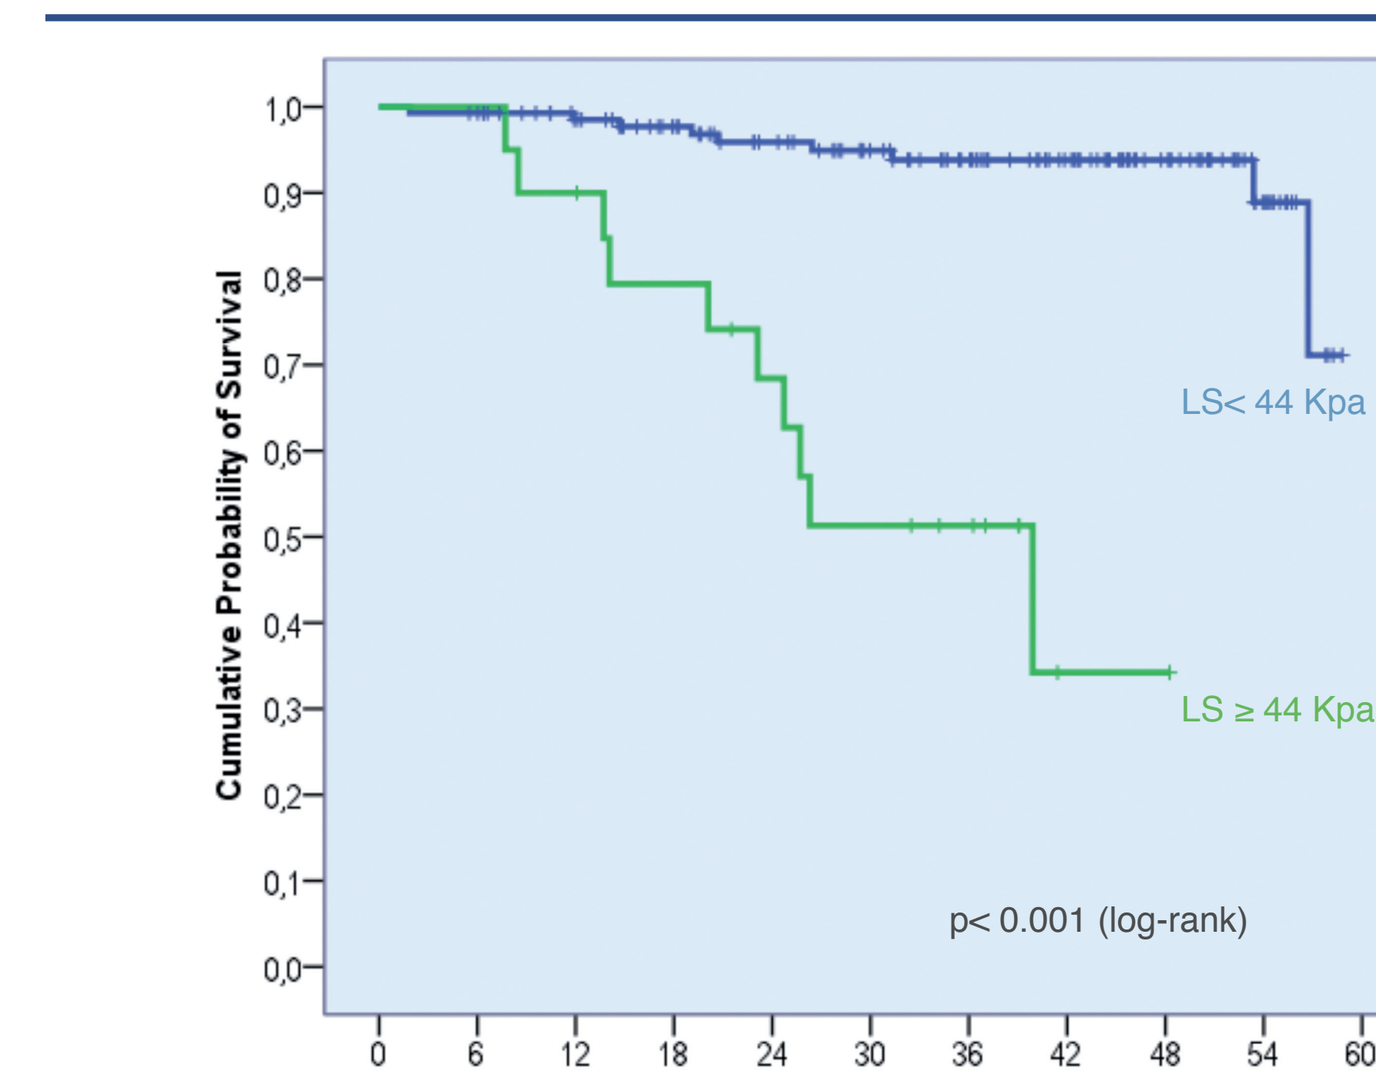
Cumulative Probability of Survival
1,0
0,9
0,8
0,7
0,6
0,5
0,4
0,3
0,2
0,1
0,0
LS< 44 Kpa
LS ≥ 44 Kpa
p< 0.001 (log-rank)
0 6 12 18 24 30 36 42 48 54 60
Months

LS< 44 Kpa	N	140	138	126	113	101	88	76	59	38	15
LS > 44 Kpa		20	20	18	15	1 2	9	7	1	1	-

COMBINED ENDPONT BY HEPATIC DECOMPENSATION AND LS MESAUREMENT

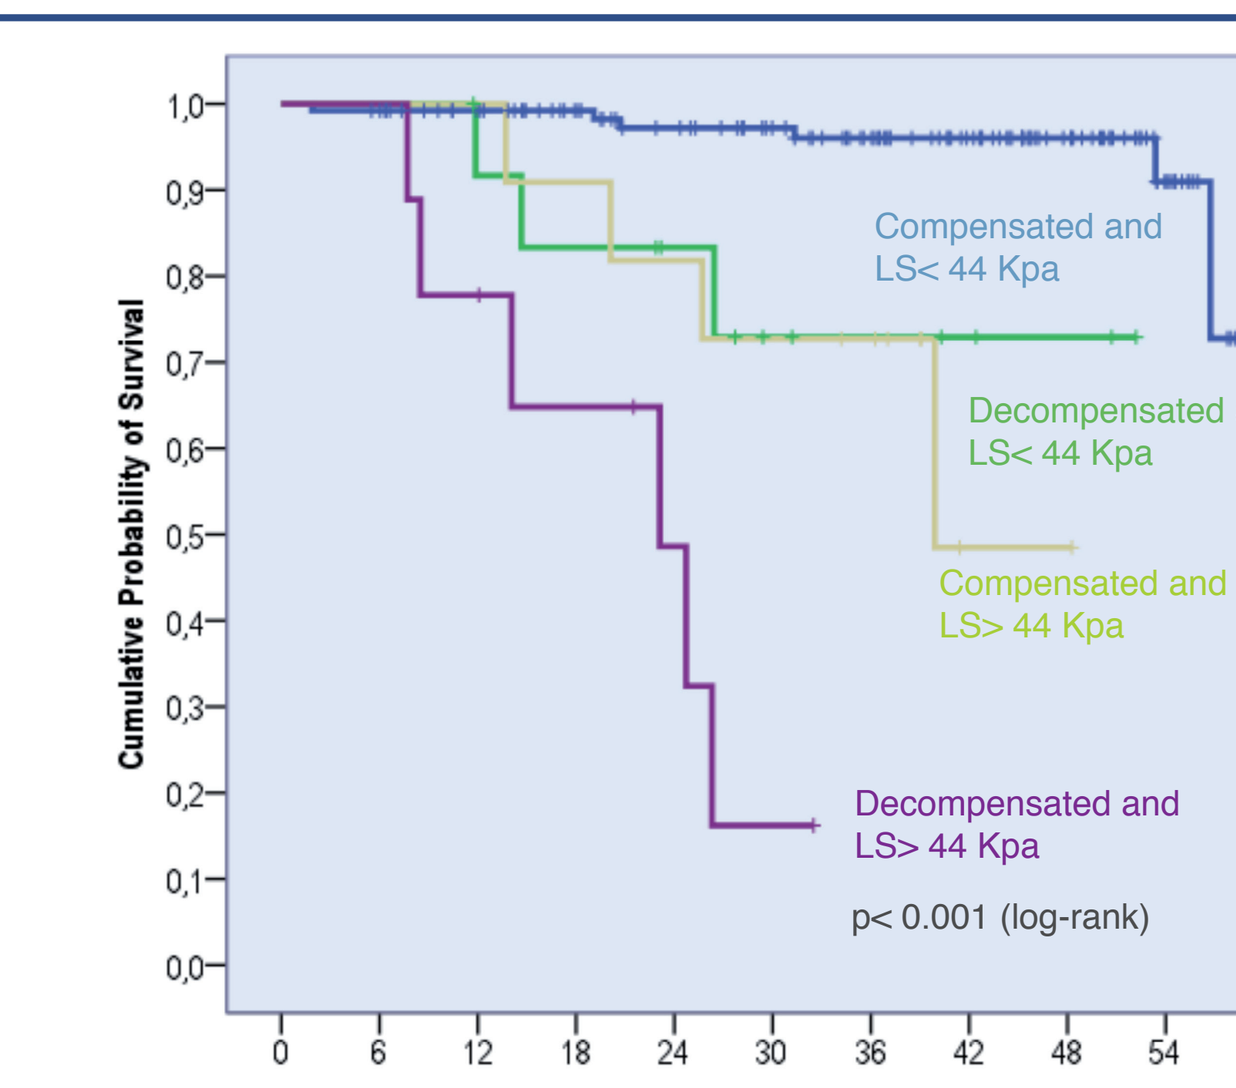
Cumulative Probability of Survival
1,0
0,9
0,8
0,7
0,6
0,5
0,4
0,3
0,2
0,1
0,0
Compensated and LS< 44 Kpa
Decompensated and LS< 44 Kpa
Compensated and LS> 44 Kpa
Decompensated and LS> 44 Kpa
p< 0.001 (log-rank)
0 6 12 18 24 30 36 42 48 54 60
Months

Compensated and LS< 44 Kpa	N	127	125	115	103	93	83	72	56	36	15	-
Decompensated and LS< 44 Kpa	N	13	13	11	10	8	5	4	3	2	-	-
Compensated and LS> 44 Kpa	N	11	11	11	10	9	8	7	1	1	-	-
Decompensated and LS> 44 Kpa	N	9	9	7	5	3	1	-	-	-	-	-

TE MEASUREMENT DIAGNOSIS PERFORMANCE

Table 6

	LS >44 for combined endpoint	LS >44 for death
AUROC	0.85 (0.76-0.94)	0.91 (0.85-0.96)
SENSITIVITY	52.6% (28.9-75.6)	64.3% (35.1-87.2)
SPECIFICITY	92.9% (87.3-96.6)	92.5% (86.9-96.2)
POSITIVE PREDICITVE VALUE	50% (27.2-72.8)	45% (23.1-68.5)
NEGATIVE PREDICTIVE VALUE	93.5 (88.2-97.0)	96.4 (91.9-98.8)
POSITIVE LIKELYHOOD RATIO	7.4 (3.2-5.5)	8.6 (3.8-15.2)
NEGATIVE LIKELYHOOD RATIO	0.5 (0.3-0.8)	0.4 (0.16-0.69)

AUROC FOR COMBINED ENDPOINT

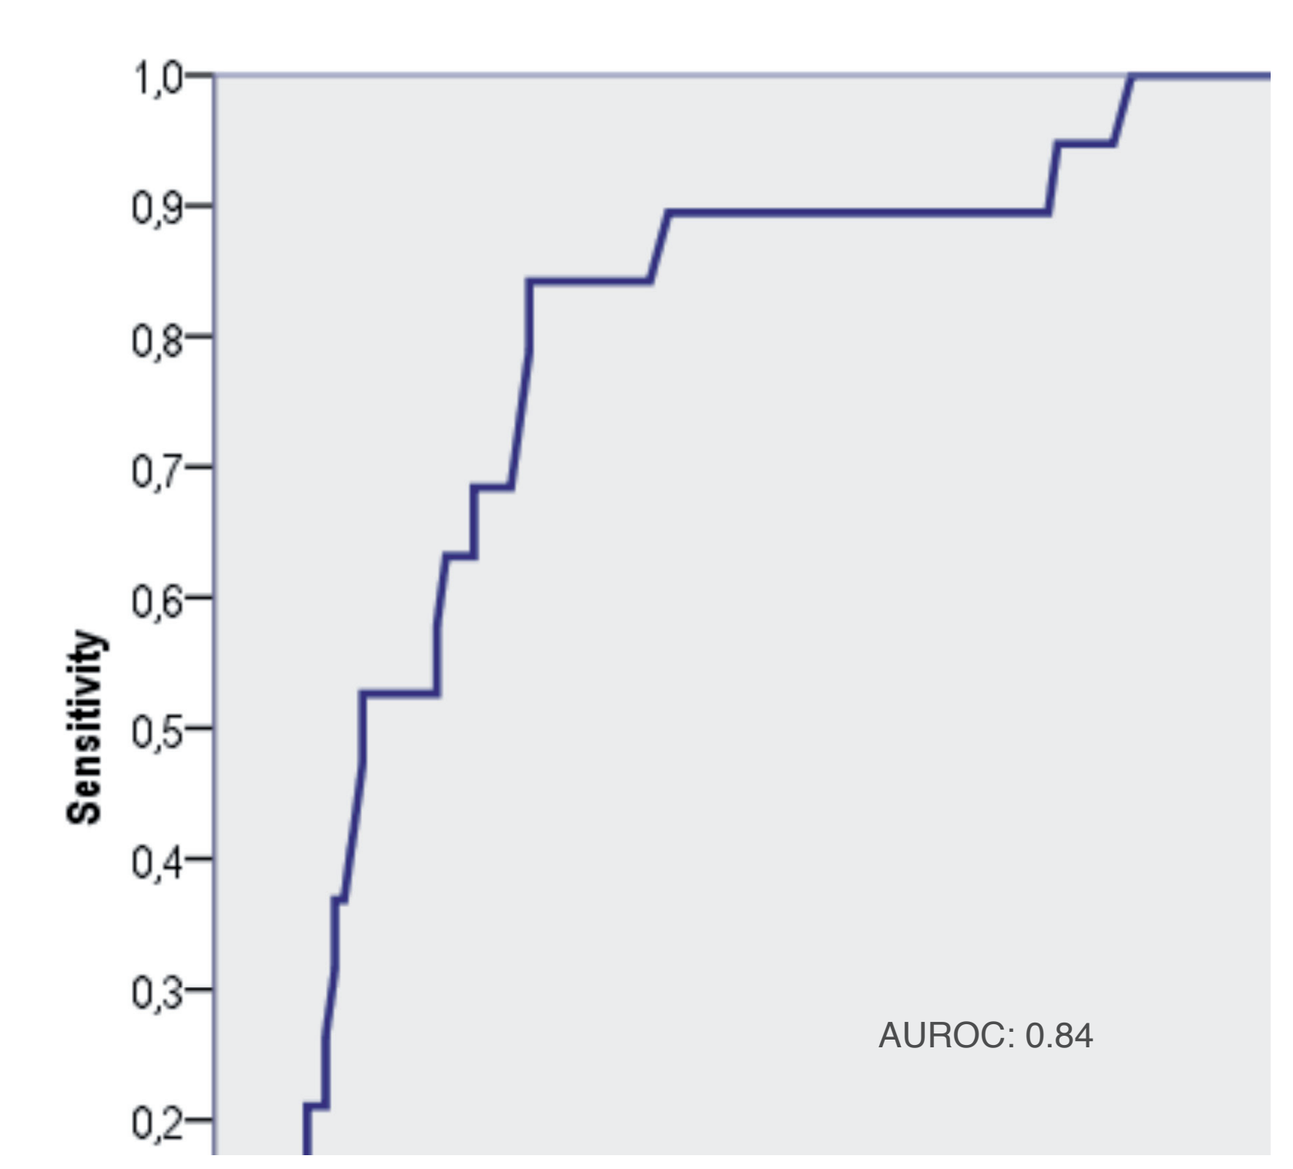
Sensitivity
1,0
0,9
0,8
0,7
0,6
0,5
0,4
0,3
0,2
AUROC: 0.84

AUROC FOR DEATH

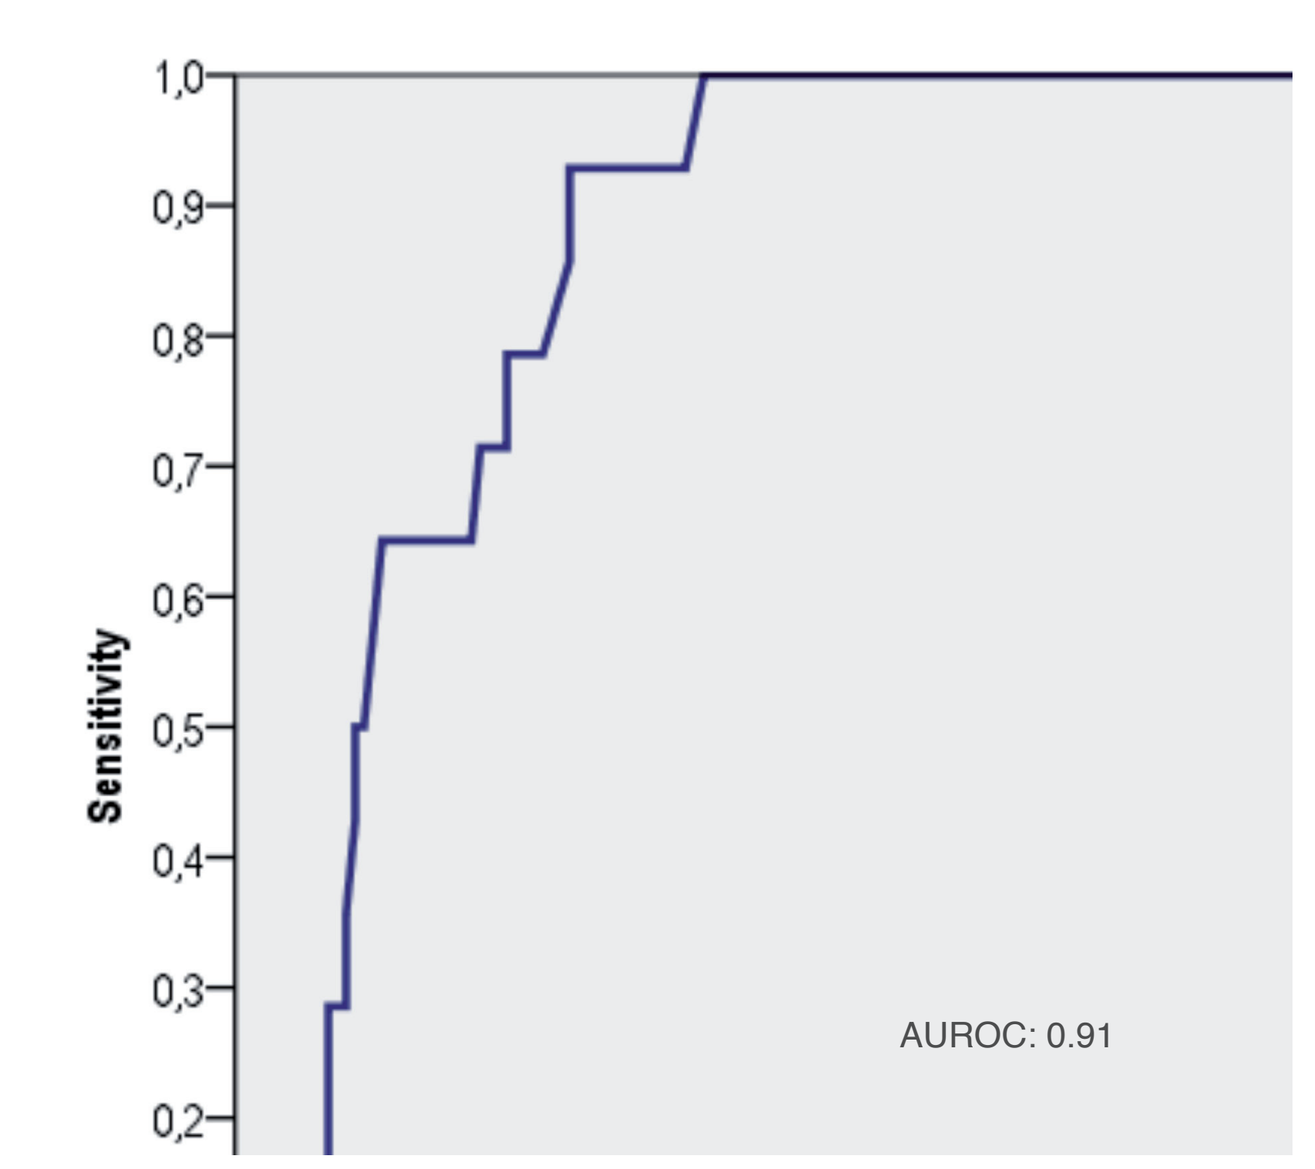
Sensitivity
1,0
0,9
0,8
0,7
0,6
0,5
0,4
0,3
0,2
AUROC: 0.91

CONCLUSIONS

- LS measurement accurately predicts survival, risk for development of hepatocarcinoma and/or liver transplantation in HIV infected subjects with liver cirrhosis.
- LS measurement predict hepatic decompensation.
- LS measurement appears to be the best prognostic test for predicting ESLD events in HIV-infected subjects with liver cirrhosis.

DISCUSSION

- Our study has some limitations:
 - » Low number of clinical events
 - » Follow-up not long enough
- These limitations might preclude showing a possible survival benefit for coinfected patients with liver cirrhosis who achieve SVR.
- Every effort should be made to identify progression of cirrhosis in HIV patients.

fipse

IdiPAZ
Instituto de Investigación
Hospital Universitario La Paz

F S G
FUNDACIÓN SEIMC-GESIDA

GESIDA 37/03-FIPSE 36465/03 -NEAT IG5 INVESTIGATORS

MªLuisa Montes Ramírez, José Ramón Arribas, José Francisco Pascual Pareja, Francisco Xavier Zamora Vargas, Juan J González-García, Juan Miguel Castro (Hospital Universitario La Paz, Madrid); Federico Pulido, María Asunción Hernando (Hospital Universitario 12 de Octubre, Madrid); Juan Berenguer Berenguer, Margarita Ramírez Schacke, Isabel Gutiérrez (Hospital General Universitario Gregorio Marañón, Madrid); Carmen Quereda, Toni Sanfrutos Alonso (Hospital Universitario Ramón y Cajal, Madrid); Alberto Arranz Caso, José Sanz-Moreno, Julio de Miguel Prieto, Esperanza Casas García (Hospital Universitario Príncipe de Asturias, Madrid). Enrique Ortega González, Laura Ortiz Ramírez de Arellano (Hospital General Universitario de Valencia), Cristina Tural Llacher, Antoni Jou Pastor (Hospital Germans Trias i Pujol, Badalona); José M. Miró, María López-Dieguez, Montse Laguno, Christian Manzardo (Hospital Clínic-IDIBAPS, Universitat de Barcelona, Barcelona). Miguel Ángel Von Wichmann de Miguel (Hospital de Donostia, San Sebastián); Herminia Esteban, Elena Barquilla (Fundación SEIMC-GESIDA, Madrid).